Poster 4095

The Safety and Efficacy of Ivonescimab in Combination With Chemotherapy as First-Line Treatment for Advanced Biliary Tract Cancer

Jieer Ying,[1] Yanru Xie,[2] Qi Xu,[1] Jingjing Li,[1] Cong Luo,[1] Lei Zhang,[1] Yonghui Wang,[2] Li Chen,[2] Jianhui Huang,[2] Wenting Li,[3] Yu Xia[3]

[1]Zhejiang Cancer Hospital, Hangzhou, China; [2]Lishui Central Hospital, Lishui, China; [3]Akeso, Inc., Zhongshan, China

BACKGROUND

- Although programmed cell death protein 1 (PD-1)/programmed death-ligand 1 (PD-L1) inhibitors combined with chemotherapy are approved as first-line treatment for advanced biliary tract cancer (BTC), the overall survival (OS) benefit is limited, particularly for patients with gall bladder cancer (GBC)[1]
- The combination of PD-1/PD-L1 and vascular endothelial growth factor (VEGF) antibodies have shown clinical benefit in patients with BTC[2,3]
- Ivonescimab is a novel, investigational bispecific antibody against PD-1 and VEGF that has exhibited cooperative binding in vitro[4] and could produce complementary and synergistic antitumor effects through both the PD-1 and VEGF pathways[4-6]
- A phase 1b/2 trial (NCT05214482) of ivonescimab plus chemotherapy with or without AK117 (an investigational antibody against CD47) for patients with advanced malignant tumors is ongoing
- Initial results from a cohort of the phase 2 portion of the study in which ivonescimab plus chemotherapy was assessed in participants with locally advanced or metastatic BTC are described herein

OBJECTIVE

- To assess the safety and preliminary antitumor activity of ivonescimab plus chemotherapy in participants with locally advanced or metastatic BTC

METHODS

Study Design

- This study includes 1 cohort of an open-label, multicenter phase 2 trial being conducted in China to evaluate ivonescimab plus chemotherapy in adults with unresectable locally advanced or metastatic intrahepatic cholangiocarcinoma (ICC), extrahepatic cholangiocarcinoma (ECC), or GBC (**Figure 1**)

Figure 1. Study Design

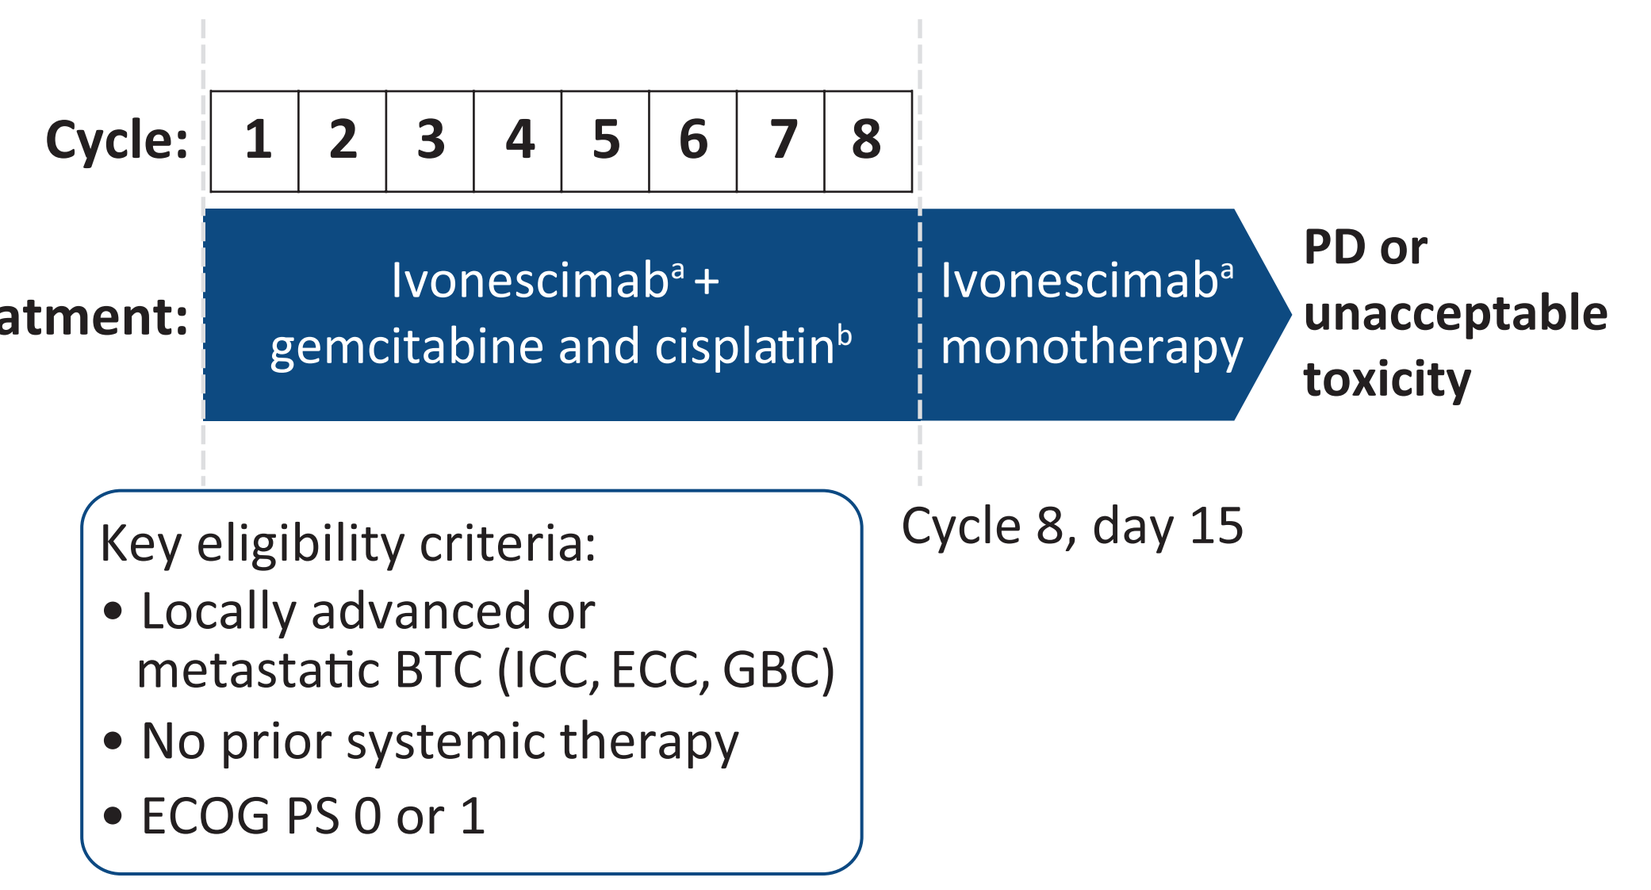
Cycle:
1 2 3 4 5 6 7 8
Treatment:
Ivonescimab[a] + gemcitabine and cisplatin[b]
Ivonescimab[a] monotherapy
PD or unacceptable toxicity
Cycle 8, day 15
Key eligibility criteria:
• Locally advanced or metastatic BTC (ICC, ECC, GBC)
• No prior systemic therapy
• ECOG PS 0 or 1

BTC, biliary tract cancer; ECC, extrahepatic cholangiocarcinoma; ECOG PS, Eastern Cooperative Oncology Group performance status; GBC, gall bladder cancer; ICC, intrahepatic cholangiocarcinoma; PD, progressive disease; Q3W, once every 3 weeks.

[a]Ivonescimab 20 mg/kg or 30 mg/kg administered Q3W on day 1 for up to 8 cycles (21 days per cycle) and administered as monotherapy Q3W until PD or unacceptable toxicity.

[b]Gemcitabine 1000 mg/m² and cisplatin 25 mg/m² administered Q3W on day 1 and day 8 for up to 8 cycles.

Outcome Measures

- Primary end points were safety and investigator-assessed objective response rate (ORR) based on Response Evaluation Criteria in Solid Tumors, version 1.1
- Secondary end points included disease control rate (DCR), progression-free survival (PFS), and OS

RESULTS

Participants

- As of January 31, 2024, a total of 22 participants with advanced BTC had enrolled in the study: 12 patients (54.5%) with ICC, 1 (4.5%) with ECC, and 9 (40.9%) with GBC (**Table 1**)
 - › At the time of data analysis, 13 participants remained in the study and 5 were still receiving the study treatment
- The median age of participants was 65.3 years (range, 47.7-75.6 years) and 81.8% of the patients (18/22) had an Eastern Cooperative Oncology Group performance status score of 1 at baseline (**Table 1**)
- The median duration of follow-up was 13.8 months, and median duration of exposure to ivonescimab was 8.3 months (interquartile range, 6.2-8.6 months)

Table 1. Baseline Characteristics

	Ivonescimab + chemotherapy N = 22
Dosing groups	
Ivonescimab 20 mg/kg + chemotherapy	13 (59.0)
Ivonescimab 30 mg/kg + chemotherapy	9 (41.0)
Age, median (range), years	65.3 (47.7-75.6)
Age <65 years	11 (50.0)
Sex	
Male	7 (31.8)
Female	15 (68.2)
ECOG PS score	
0	4 (18.2)
1	18 (81.8)
Primary tumor type	
ICC	12 (54.5)
ECC	1 (4.5)
GBC	9 (40.9)
Disease classification	
Locally advanced	3 (13.6)
Metastatic	19 (86.4)
PD-L1 combined positive score	
<1	6 (27.3)
≥1	10 (45.5)
Unknown	6 (27.3)

ECC, extrahepatic cholangiocarcinoma; ECOG PS, Eastern Cooperative Oncology Group performance status; GBC, gall bladder cancer; ICC, intrahepatic cholangiocarcinoma; PD-L1, programmed death-ligand 1.

Values are No. (%) unless otherwise noted.

Race/ethnicity demographics data were not collected.

Safety

- Grade ≥3 treatment-related adverse events (TRAEs) were reported in 86.4% of participants, but none led to discontinuation of treatment or to death (**Table 2**)
- The most common grade 3 or 4 TRAEs, with a frequency of at least 10%, were anemia (40.9%), white blood cell count decreased (36.4%), neutrophil count decreased (36.4%), platelet count decreased (18.2%), and hypertension (18.2%; **Figure 2**)

Table 2. Summary of Safety Results

	Ivonescimab + chemotherapy N = 22
TEAEs	
Grade ≥3	21 (95.5)
Serious TEAEs	15 (68.2)
Leading to discontinuation	0
Leading to death	0
TRAEs	
Grade ≥3	19 (86.4)
Treatment-related serious AEs	8 (36.4)
Leading to discontinuation	0
Leading to death	0
Immune-related AEs	7 (31.8)
Grade ≥3	4 (18.2)
Gamma-glutamyl transferase increased	1 (4.5)
Lipase increased	1 (4.5)
Platelet count decreased	1 (4.5)
Rash	1 (4.5)

AE, adverse event; TEAE, treatment-emergent adverse event; TRAE, treatment-related adverse event.

Values are No. (%) unless otherwise noted.

Figure 2. Most Common (≥10%) TRAEs

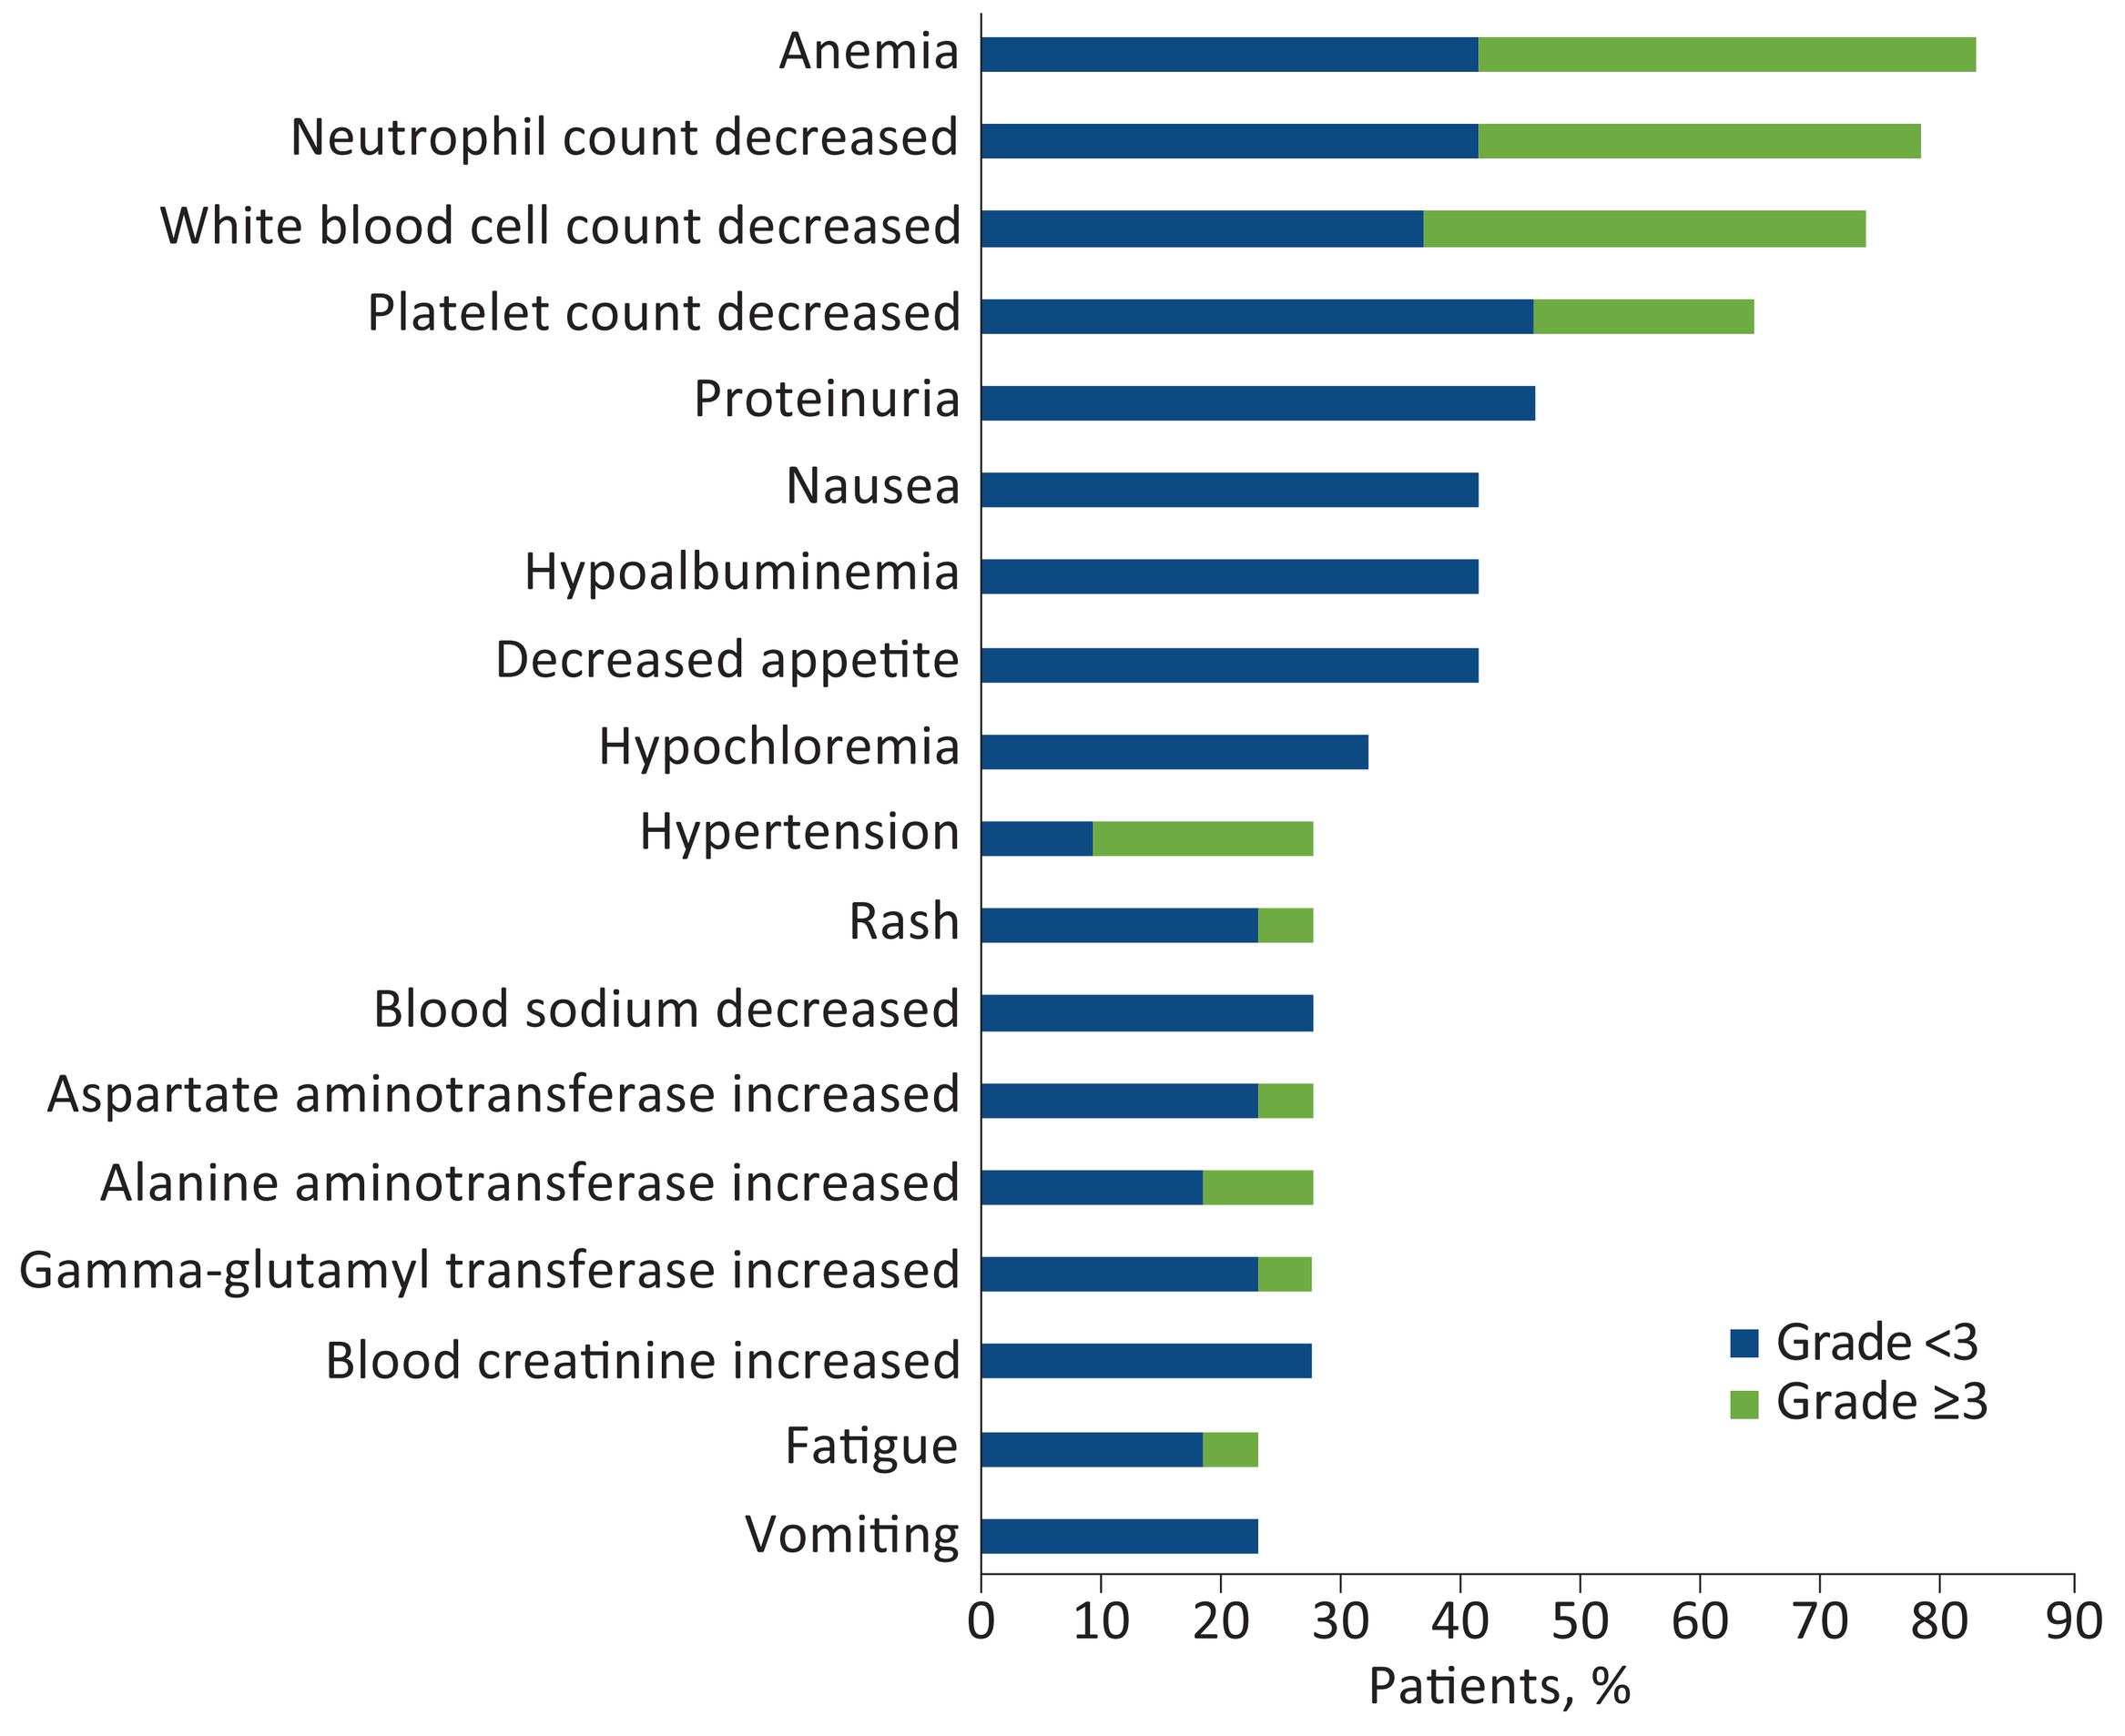
Anemia
Neutrophil count decreased
White blood cell count decreased
Platelet count decreased
Proteinuria
Nausea
Hypoalbuminemia
Decreased appetite
Hypochloremia
Hypertension
Rash
Blood sodium decreased
Aspartate aminotransferase increased
Alanine aminotransferase increased
Gamma-glutamyl transferase increased
Blood creatinine increased
Fatigue
Vomiting
0 10 20 30 40 50 60 70 80 90
Patients, %
Grade <3
Grade ≥3

TRAE, treatment-related adverse event.

Efficacy

- ORR by investigator assessment was 63.6% (14/22) overall (**Table 3**) and was 77.8% (7/9) for participants with GBC
 - › Results were consistent between the ivonescimab 20 mg/kg and 30 mg/kg Q3W dosing groups
 - › Individual patient-level response is shown in **Figures 3** and **4**
- DCR was 100% (22/22, 95% confidence interval [CI], 84.6-100)

Table 3. Overview of Objective Response Rate

	Ivonescimab + chemotherapy N = 22
Patients who had an objective response, No.	14
ORR (95% CI), %	63.6 (40.7-82.8)
Best overall response, No. (%)	
CR	0
PR	14 (63.6)
SD	8 (36.4)
PD	0

CR, complete response; ORR, objective response rate; PD, progressive disease; PR, partial response; SD, stable disease

Figure 3. Best Percentage Change From Baseline in Sum of Tumor Diameters

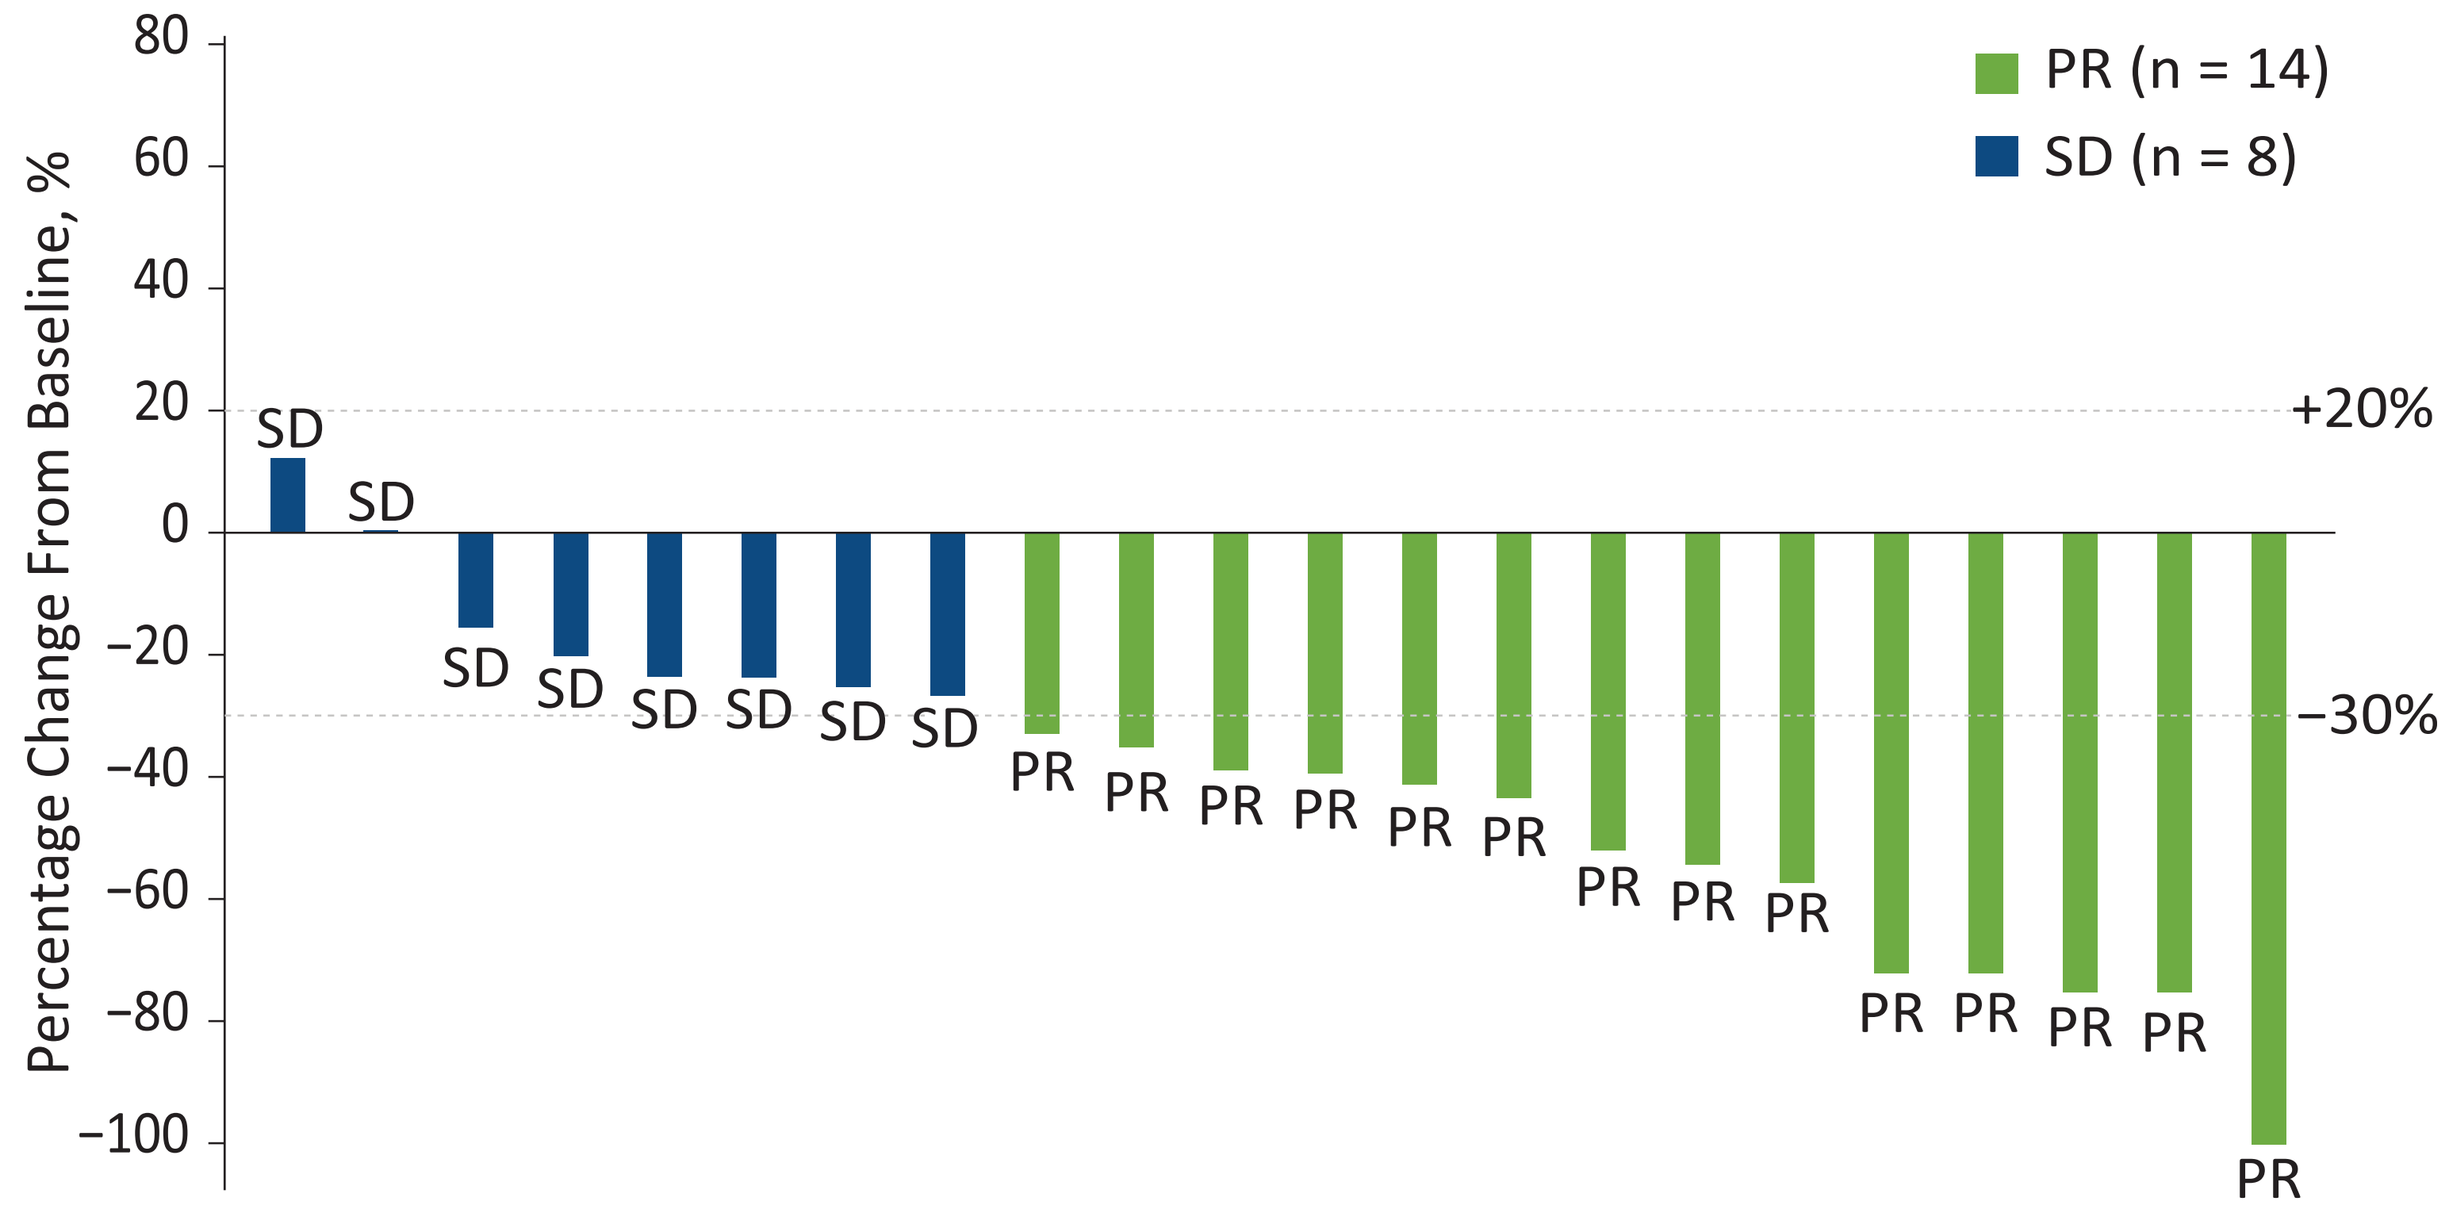
Percentage Change From Baseline, %
PR (n = 14)
SD (n = 8)
+20%
−30%

n, number of participants; PR, partial response; SD, stable disease.

Figure 4. Percentage Change from Baseline in Sum of Diameters Over Time

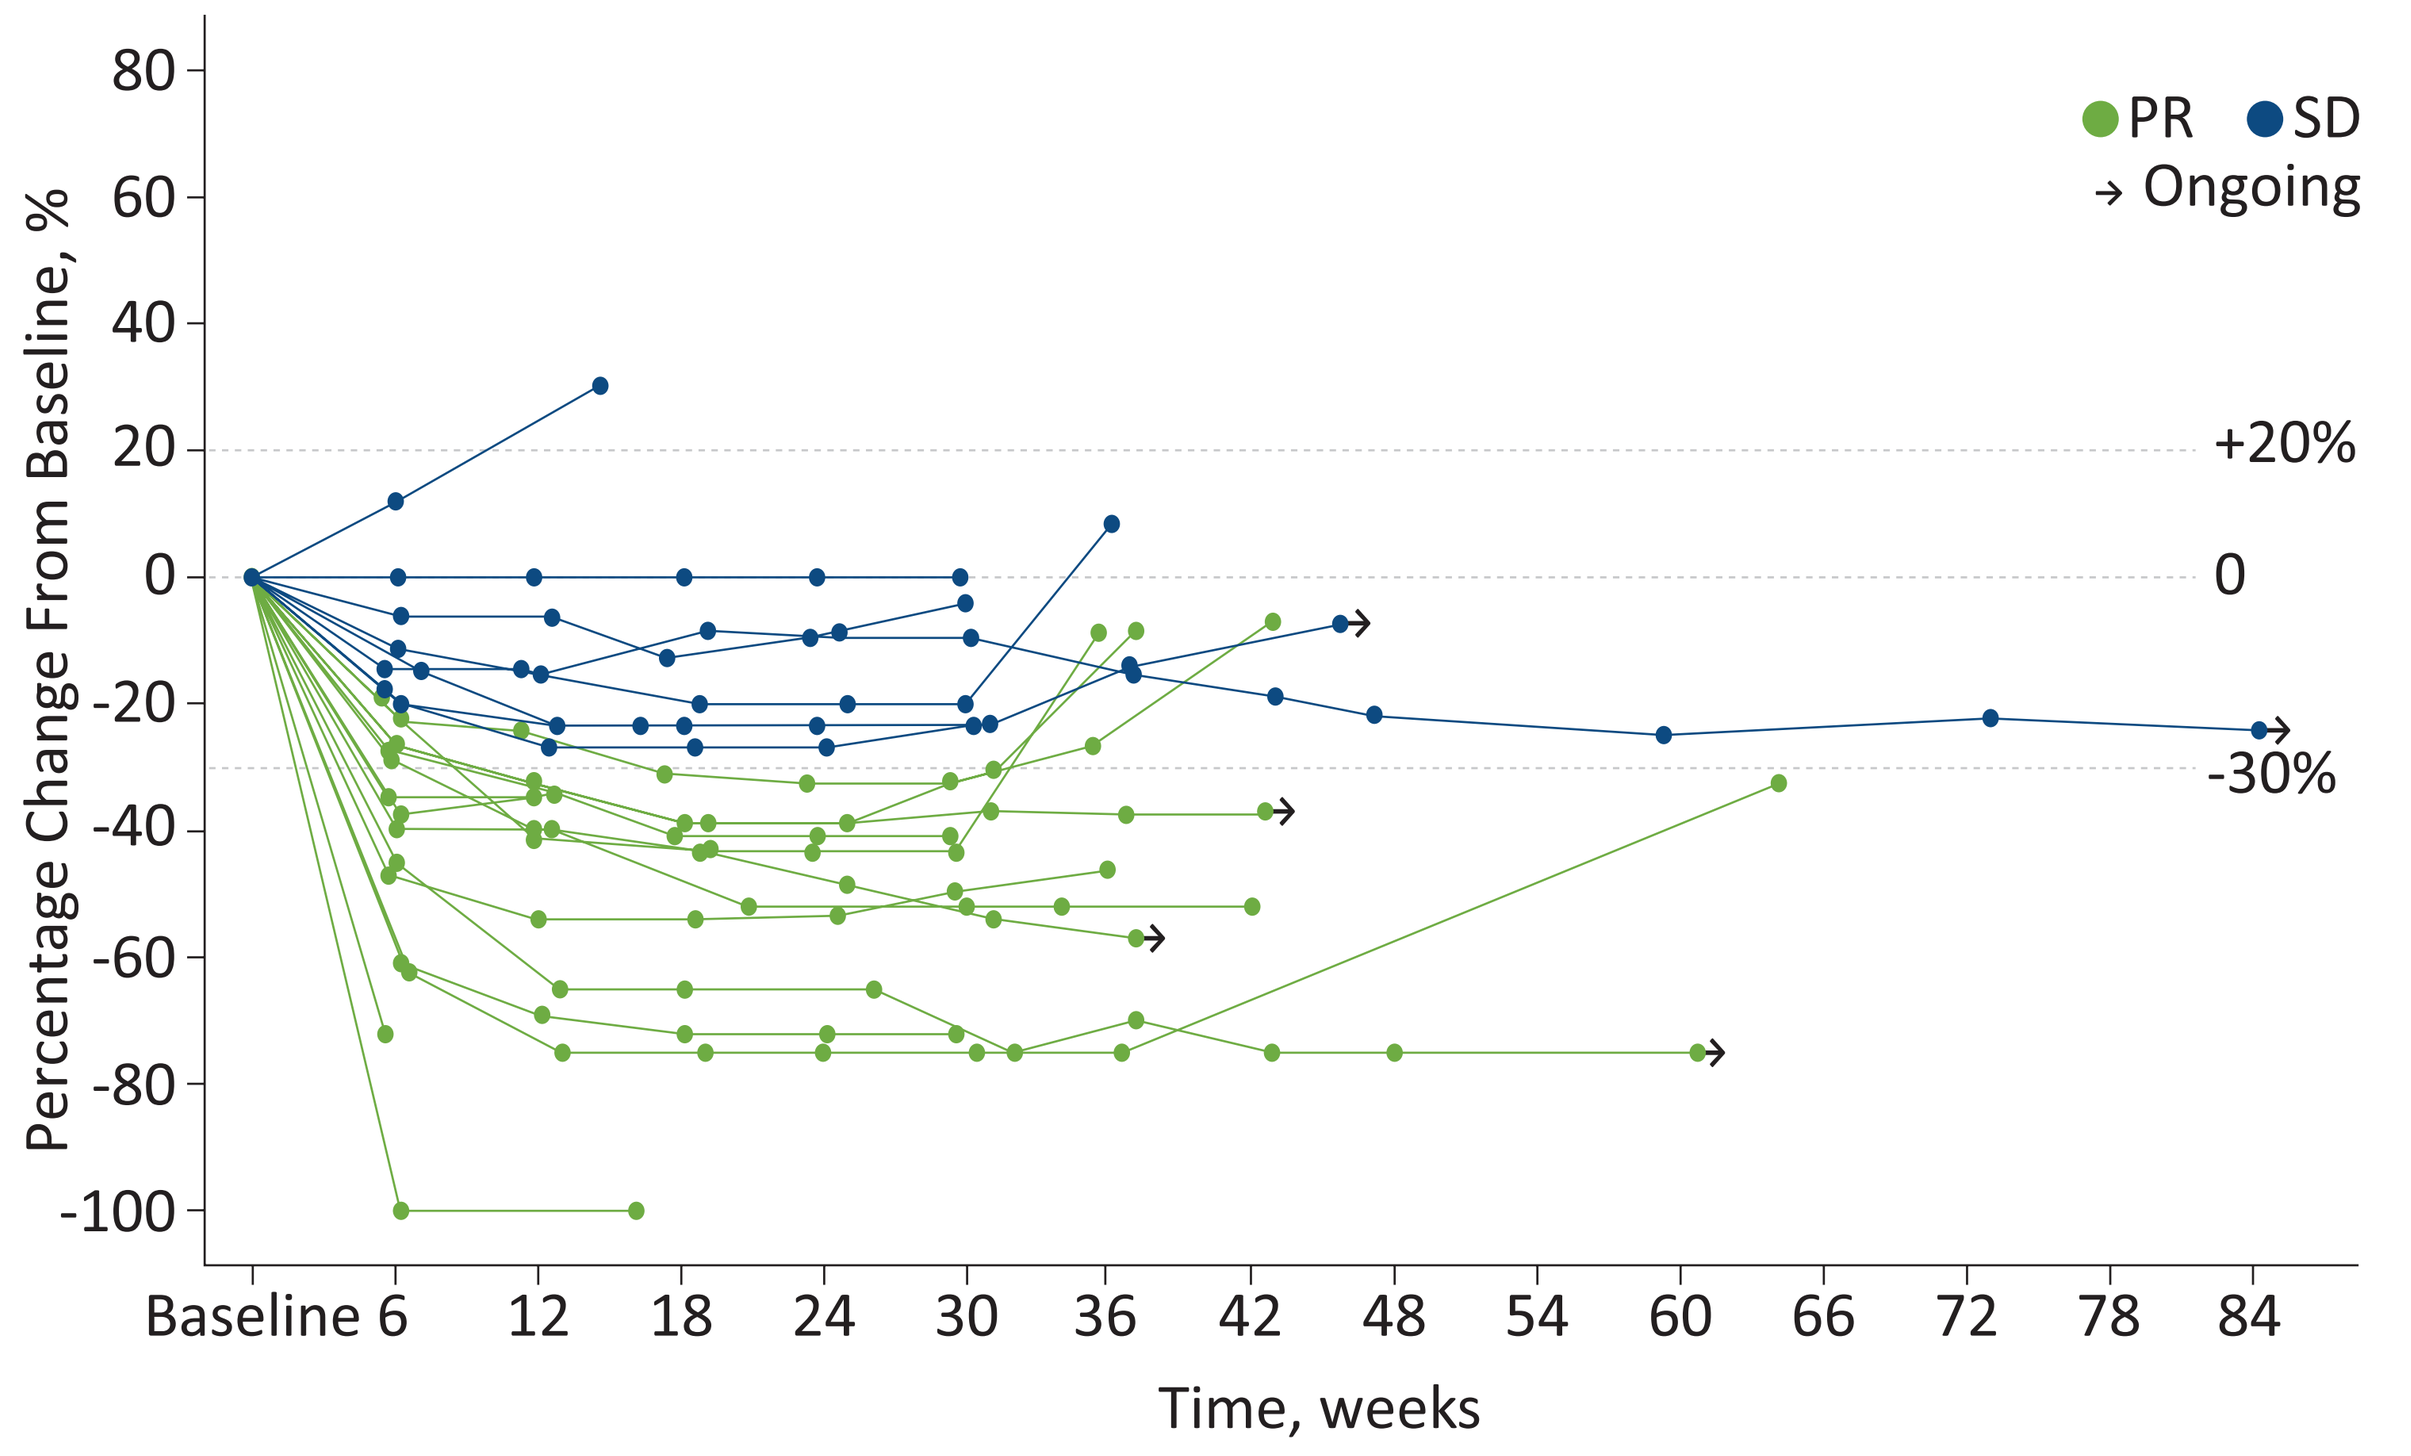
Percentage Change From Baseline, %
PR SD
→ Ongoing
+20%
0
-30%
Time, weeks

PR, partial response; SD, stable disease.

- The median PFS was 8.5 months (95% CI, 6.8-10.5), with a 6-month PFS rate of 84.4% (95% CI, 59.1-94.7; **Figure 5**)
- The median OS was 16.8 months (95% CI, 9.2 to not estimable [NE]), with a 9-month OS rate of 81.8% (95% CI, 58.5-92.8; **Figure 6**)
 - › In patients with GBC, the median OS was 16.8 months (95% CI, 3.8-NE)

Figure 5. Kaplan-Meier Curve of Progression-Free Survival

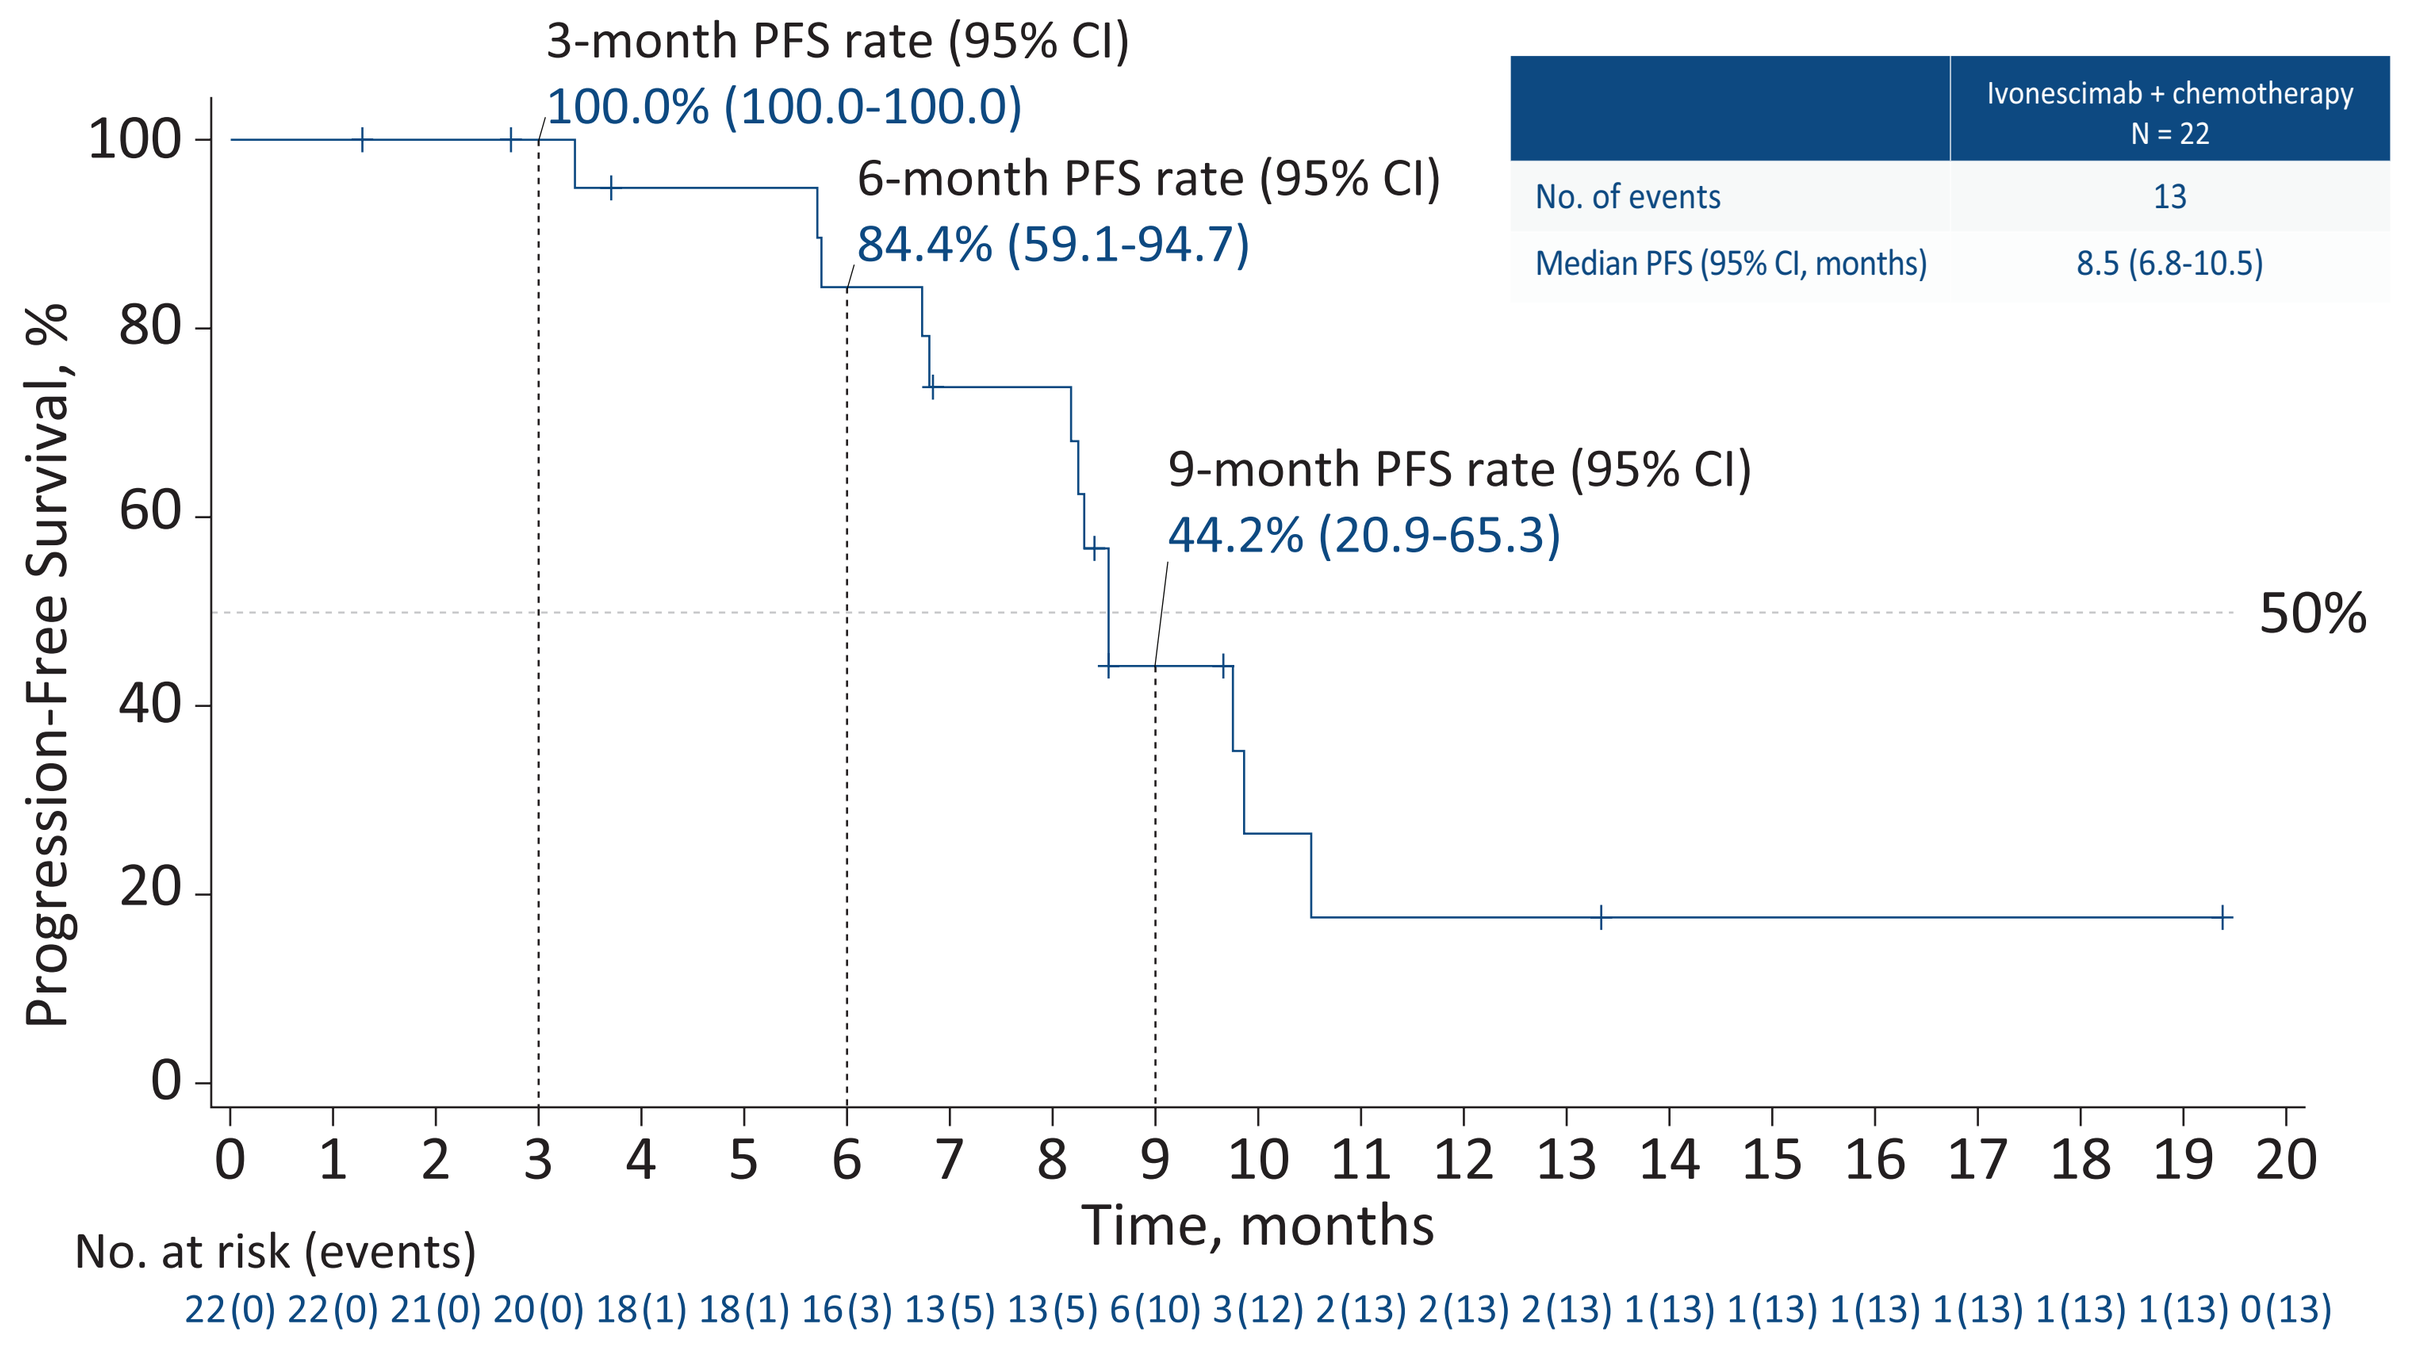
3-month PFS rate (95% CI)
100.0% (100.0-100.0)
6-month PFS rate (95% CI)
84.4% (59.1-94.7)
9-month PFS rate (95% CI)
44.2% (20.9-65.3)
50%
Ivonescimab + chemotherapy N = 22
No. of events 13
Median PFS (95% CI, months) 8.5 (6.8-10.5)
Progression-Free Survival, %
Time, months
No. at risk (events)
22(0) 22(0) 21(0) 20(0) 18(1) 18(1) 16(3) 13(5) 13(5) 6(10) 3(12) 2(13) 2(13) 2(13) 1(13) 1(13) 1(13) 1(13) 1(13) 1(13) 0(13)

N, total number of participants; PFS, progression-free survival.

Figure 6. Kaplan-Meier Curve of Overall Survival

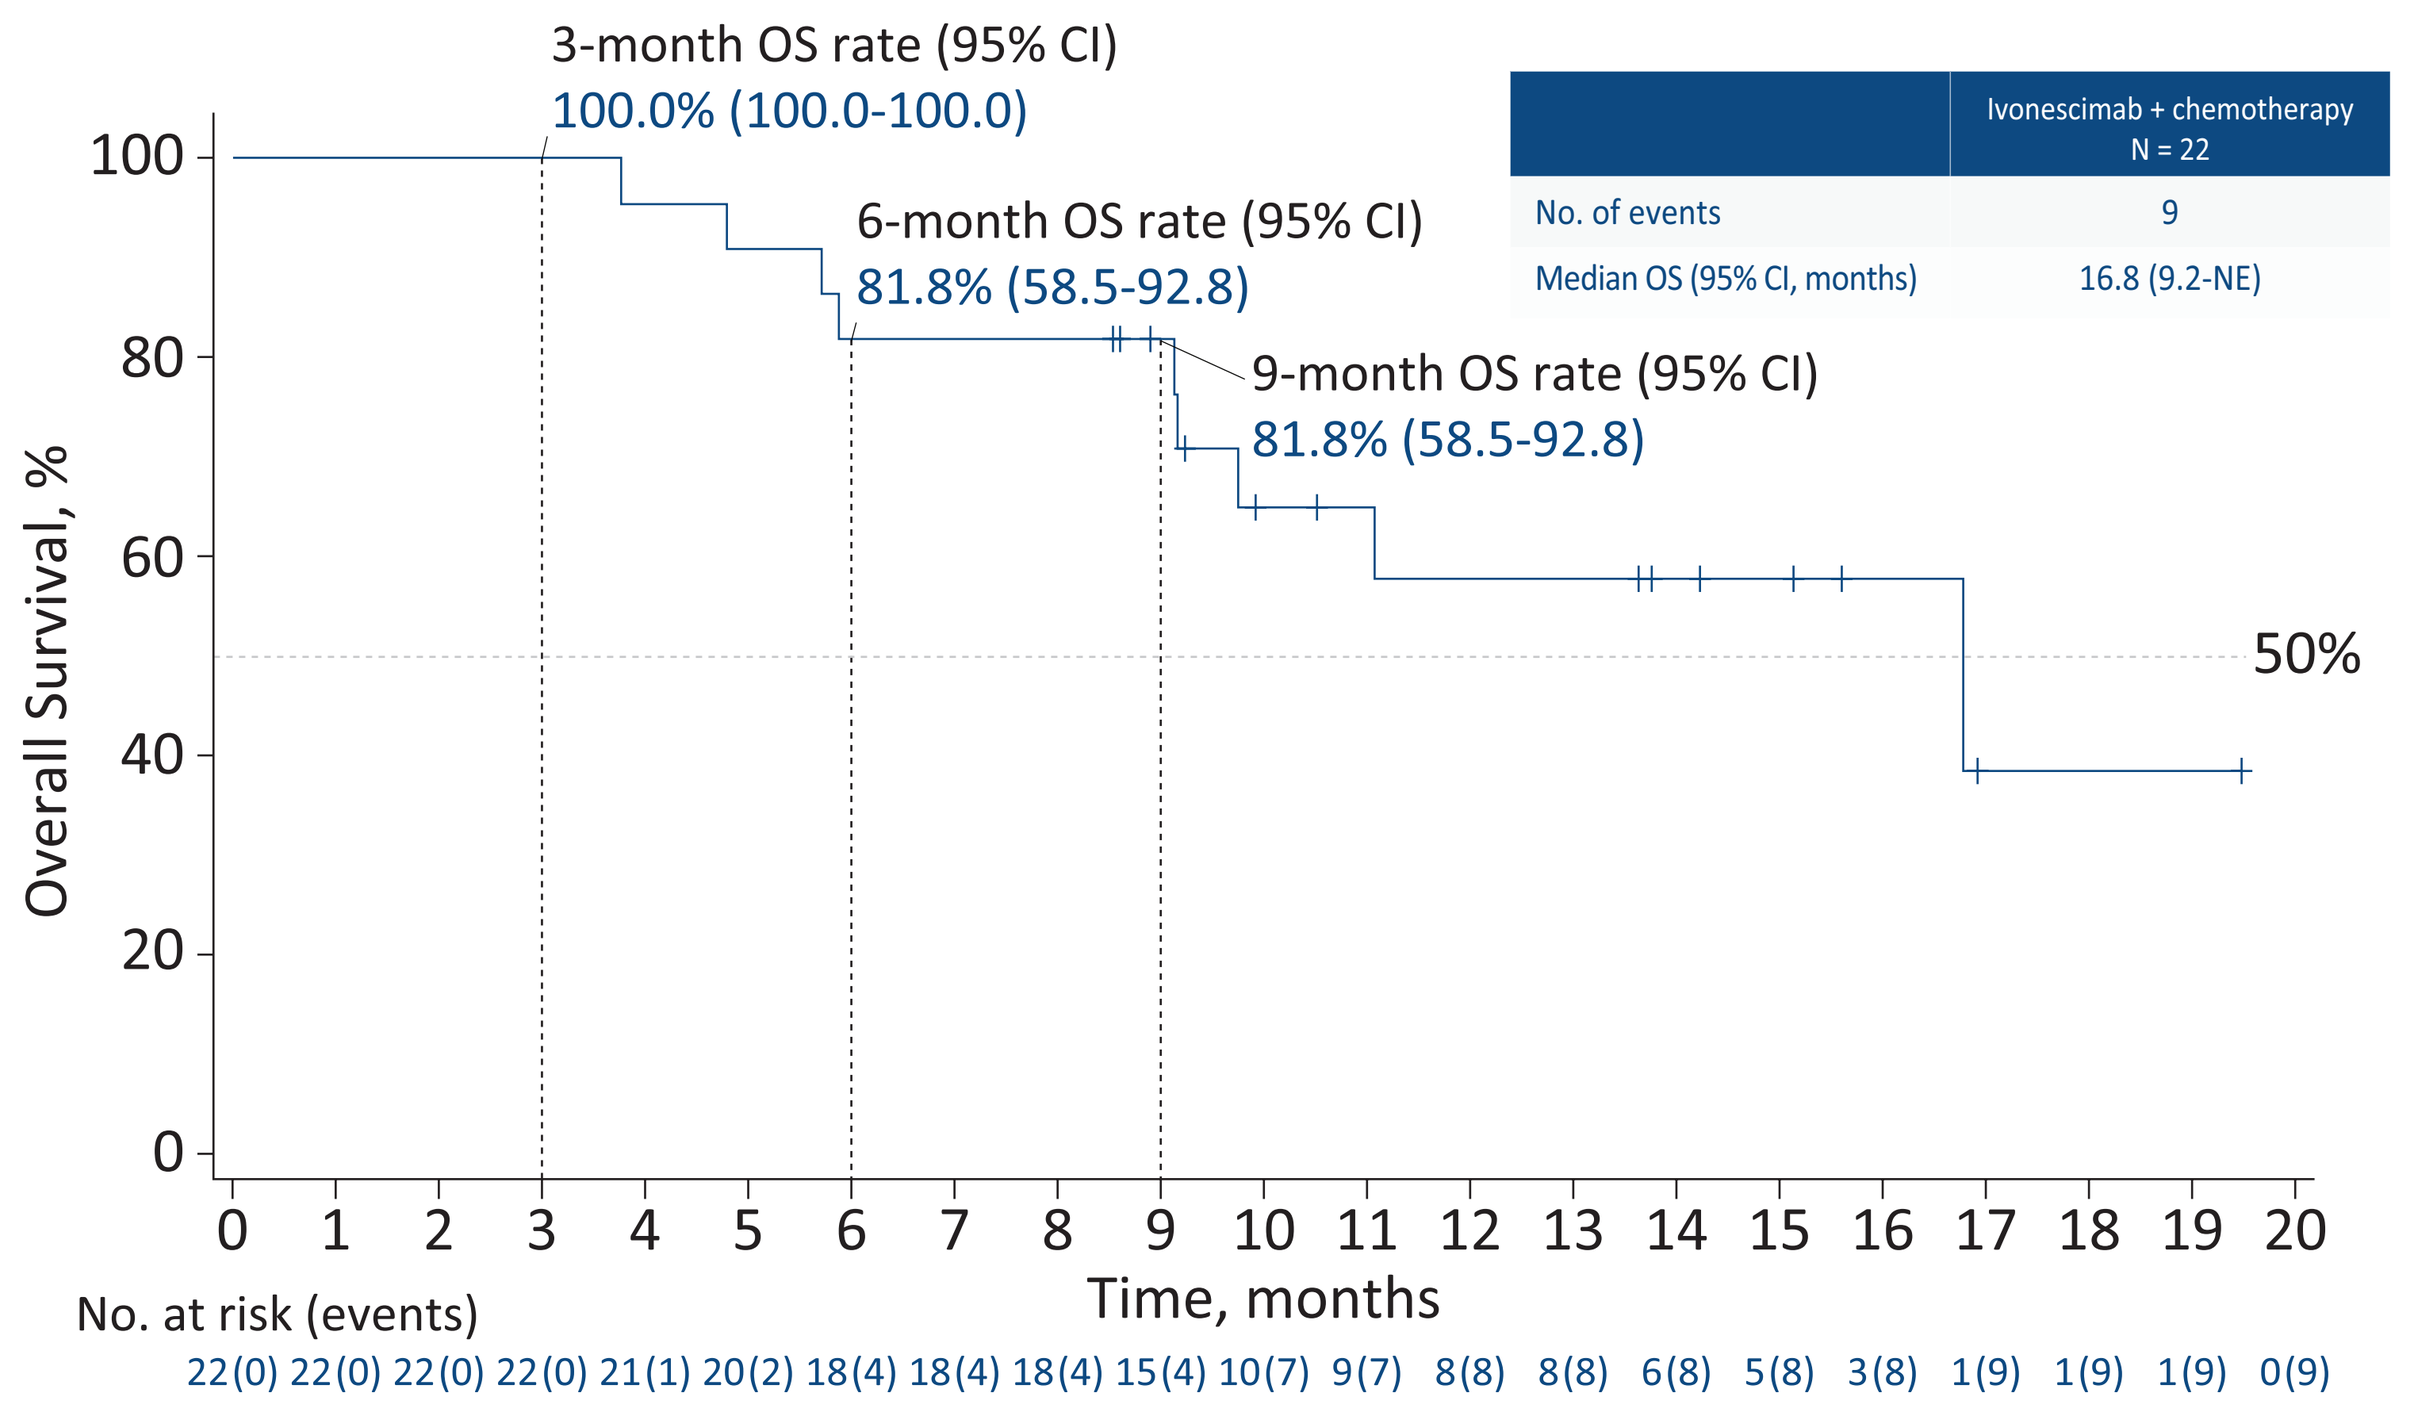
3-month OS rate (95% CI)
100.0% (100.0-100.0)
6-month OS rate (95% CI)
81.8% (58.5-92.8)
9-month OS rate (95% CI)
81.8% (58.5-92.8)
50%
Ivonescimab + chemotherapy N = 22
No. of events 9
Median OS (95% CI, months) 16.8 (9.2-NE)
Overall Survival, %
Time, months
No. at risk (events)
22(0) 22(0) 22(0) 22(0) 21(1) 20(2) 18(4) 18(4) 18(4) 15(4) 10(7) 9(7) 8(8) 8(8) 6(8) 5(8) 3(8) 1(9) 1(9) 1(9) 0(9)

N, total number of participants; NE, not estimable; OS, overall survival.

CONCLUSIONS

- Ivonescimab (20 or 30 mg/kg Q3W) combined with chemotherapy showed promising antitumor activity and an acceptable safety profile in participants with unresectable locally advanced or metastatic BTC
 - › ORRs were comparable across dose groups in this preliminary analysis
- The most frequently reported grade 3 or 4 TRAEs (≥10%) were anemia, white blood cell count decreased, neutrophil count decreased, platelet count decreased, and hypertension
- Results of this analysis support further evaluation of ivonescimab plus chemotherapy for the treatment of BTC
- Ivonescimab is an investigational therapy that is not approved by any regulatory authority

ACKNOWLEDGMENTS

The authors thank the participants and their caregivers, the investigators, and all the site staff who have been participating in this study. This study is sponsored by Akeso Biopharma, Inc. Medical writing assistance was provided by ApotheCom (Yardley, PA, USA).

REFERENCES

1. Lo JH et al. *Cancers (Basel)*. 2023;15:3312.
2. Iyer RV et al. *Am J Clin Oncol.* 2018;41:649-655.
3. Arkenau HT et al. *Oncologist.* 2018;23:1407-e136.
4. Zhong T et al. Presented at AACR-NCI-EORTC International Conference; October 11-15, 2023; Boston, Massachusetts; Poster #B123, abstract #35333.
5. Zhao Y et al. *EClinicalMedicine.* 2023;62:102106.
6. Wang L et al. *J Thorac Oncol.* 2024;19:465-474.

DISCLOSURES

Wenting Li and Yu Xia are employees of Akeso Biopharma, Inc.

Please contact the author at hzyingjieer@163.com for questions or comments.

Copies of this poster obtained through Quick Response (QR) Code are for personal use and may not be reproduced without permission from ASCO® or the authors of this poster. By use of the QR Code you agree to Summit's privacy notice located at smmttx.com.

Presented at the American Society of Clinical Oncology (ASCO) Annual Meeting; May 31-June 4, 2024; Chicago, Illinois